Pediatric Organ Donor Outcomes an Observational Study

C Yancey, R Ketcham, A Milam, B Nave, C Hostetler, JP Orlowski
LifeShare Network, Inc., Oklahoma City, Oklahoma

Purpose

To study the outcomes of a donor hospital with an intensive care unit that excels in early identification and support of potential pediatric donors and optimal care throughout both ICU management and the donation process if family authorizes for donation with a high level of consistency.

Method

Retrospective case series study evaluating donation outcomes at one donor hospital within the organ procurement organizations (OPOs) donation service area (DSA). For each donor recovered at this donor hospital the Scientific Registry for Transplant Recipients (SRTR) observed versus expected (O:E) calculation was performed and reported.

Results

The donor hospital demonstrated consistently better than expected organs transplanted vs expected for all cases recovered, authorized, and facilitated at this donor hospital across the entire case series. The outcomes (Table 1) demonstrated an O:E ranging from a low of 1.04 to a high of 1.50. with an average of 1.20.

Table 1. O:E per case

	O:E	# transplanted	# expected	O Yield per 100 donors	E yield per 100 donors	O:E yield per 100 Donors
Case 1	1.50	6	4	600	399.4	200.6
Case 2	1.16	3	2	400	343.3	56.6
Case 3	1.16	4	3	400	343.3	56.6
Case 4	1.13	4	4	400	353.1	46.9
Case 5	1.04	4	4	400	386.4	13.6

Conclusion

This donor hospital's pediatric ICU staff, physicians and leadership has historically demonstrated a culture of support for donation. By identifying potential donor patients, making early referrals, maintaining donation through critical care management, and partnering with the OPO, the donor hospital has achieved excellent donation outcomes for patients after life-saving efforts have been exhausted. Further study is warranted to compare these donation outcomes to other pediatric donor hospitals outcomes in this DSA and against national pediatric donation outcomes. The OPO and hospital are collaborating to produce guidance and best practices for pediatric donor identification, optimal strategies for maintaining donor potential and donor management post authorization that can be used across the OPO's DSA and shared in a case series manuscript nationally.

The authors of the presentation have no conflict of interest
or financial relationships related to this abstract.