Optimizing Staffing Efficiency by Shifting Overnight Organ Referral Triage

LifeShare Network

A Harrison, B Brewer, C Yancey, C Horkey, K Gillespie, M Beeman, S LaPlante, JP Orlowski

LifeShare Network, Inc., Oklahoma City, Oklahoma

Purpose

This study evaluated the impact that cross training the Donation Services (DS) Department to perform overnight organ referrals triage had on the on-call teams at one OPO.

Methods

A retrospective review was conducted, examining the overall number of organ night referrals received by the OPO. This review examined how many of the night referrals were able to be triaged by DS staff after all DS night shift employees were cross trained on the organ referral triage process. The goal was to evaluate how many hours the cross training shifted from on call personnel to the DS during the evaluated timeframe. Triage training happened prior to August 2022. Data was reviewed from August 2022 to December 2023.

Results

The total number of organ referrals that were received, excluding any automatic rule out, was reviewed along with the number of these referrals that the DS team completed the organ triage that historically would be done by an on-call staff member. For an easy evaluation, we assumed and estimated that each triage call took one hour to complete. During this period the average number of referrals received during the night shift, 1900-0700, was 84 per month; while the average number of referrals completed by the DS team was 39. This resulted in an average of 39 hours per month being shifted from on-call personnel to on-shift personnel.

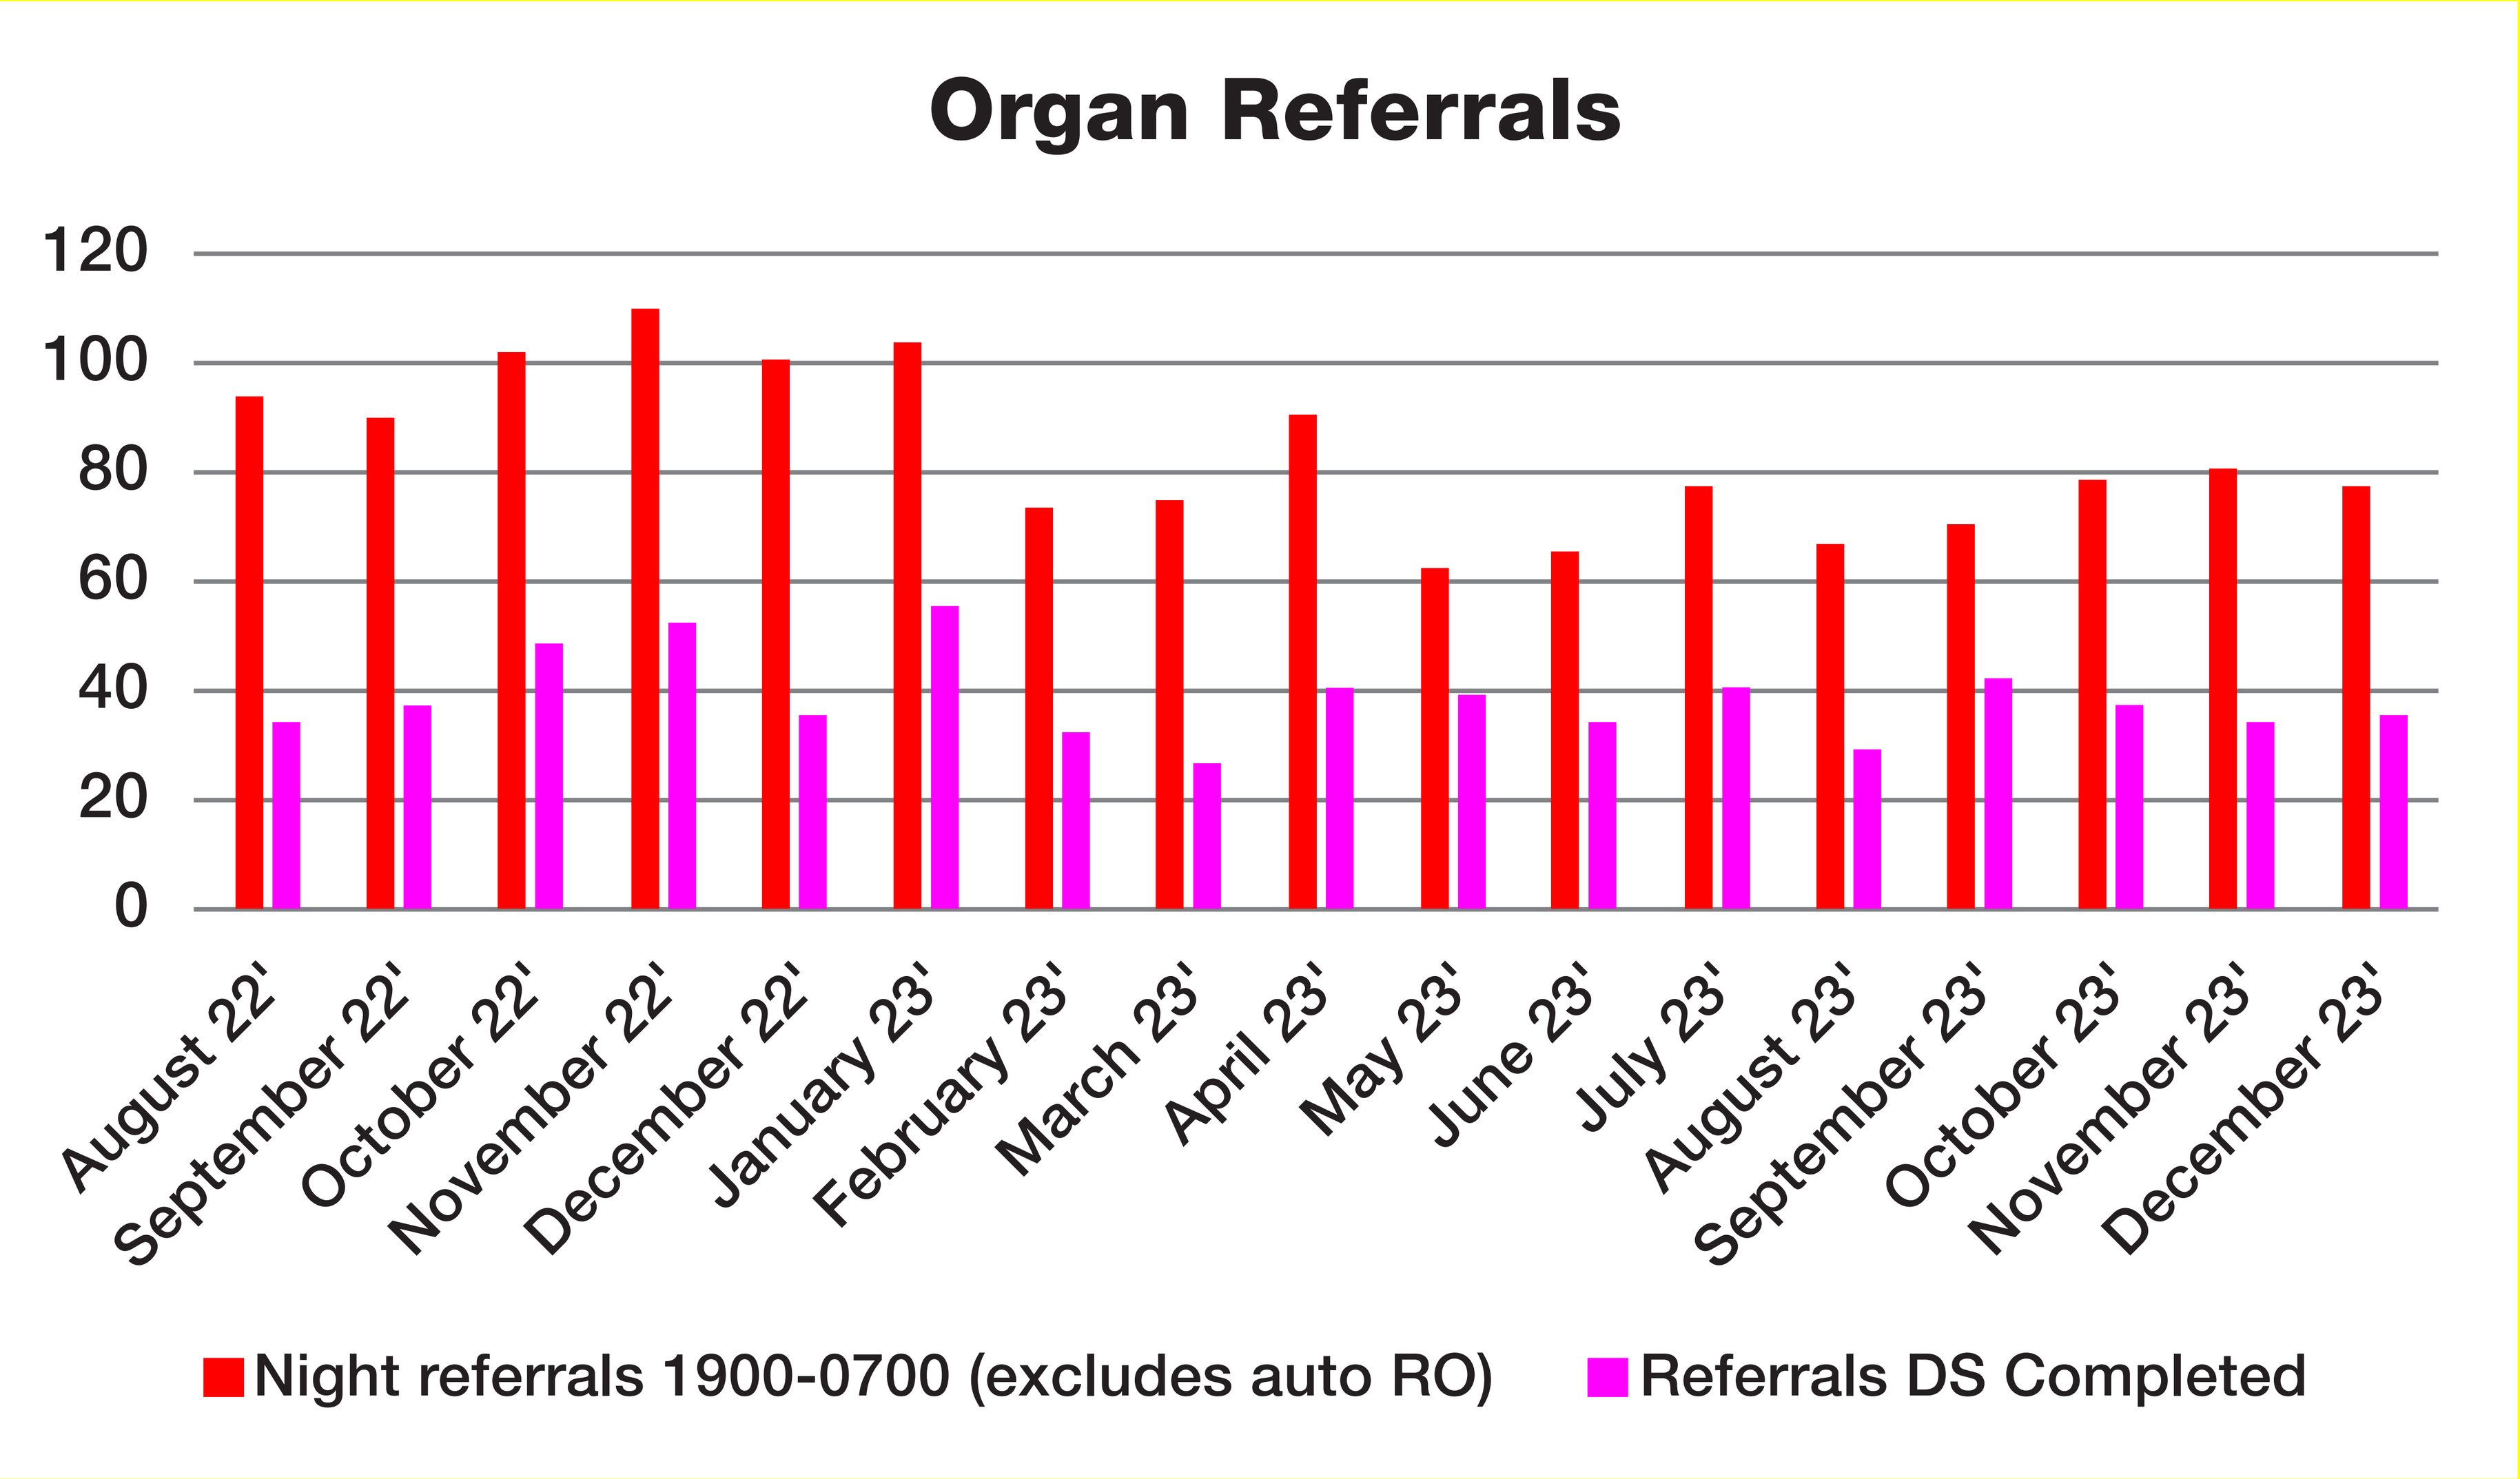

Organ Referrals
120
100
80
60
40
20
0
August 22'
September 22'
October 22'
November 22'
December 22'
January 23'
February 23'
March 23'
April 23'
May 23'
June 23'
July 23'
August 23'
September 23'
October 23'
November 23'
December 23'
Night referrals 1900-0700 (excludes auto RO)
Referrals DS Completed

Discussion

The program to train on-shift personnel to assist with overnight organ referrals has resulted in increased workforce efficiency for the organization. From a quantitative standpoint, it has resulted in a significant shift of approximately 39 working hours per month from on-call staff to on-shift staff. Beyond these measurable gains, there have been notable anecdotal improvements based on on-call staff feedback. These include a better work-life balance for on-call personnel and enabling them to dedicate more focused time to family support, approach, and authorization during regular daytime hours. The initiative has also maximized the efficiency of DS time during their working shift while fostering increased camaraderie between DS and on-call staff. Finally, it has led to a greater number of staff members being trained for a core function of the organization. In summary, the training of DS staff for overnight organ triage has brought about multifaceted benefits for the organization.

The authors of the presentation have no conflict of interest or financial relationships related to this abstract.