A Pioneering Collaboration Between OPOs Utilizing NMP

A.C. O'Neil, A.G. Johnson, A. Milam, S.F. Li, C.A. Hostetler, R.A. Squires, J.P. Orlowski

LifeShare Network

LifeShare Network, Inc.
Oklahoma City, Oklahoma

Purpose

The disparity between the demand for liver transplants and the availability of suitable donor organs is an ongoing critical issue. Normothermic machine perfusion (NMP) technology represents a pivotal advancement in organ donation, to increase utilization of available donated livers. An Organ Procurement Organization (OPO) regional-based perfusion model has potential to increase use and efficiently expand availability of this technology. This abstract explores a proof-of-concept case describing the synergistic approach of employing normothermic perfusion through regional OPO collaboration as a model to enhance liver utilization rates nationally.

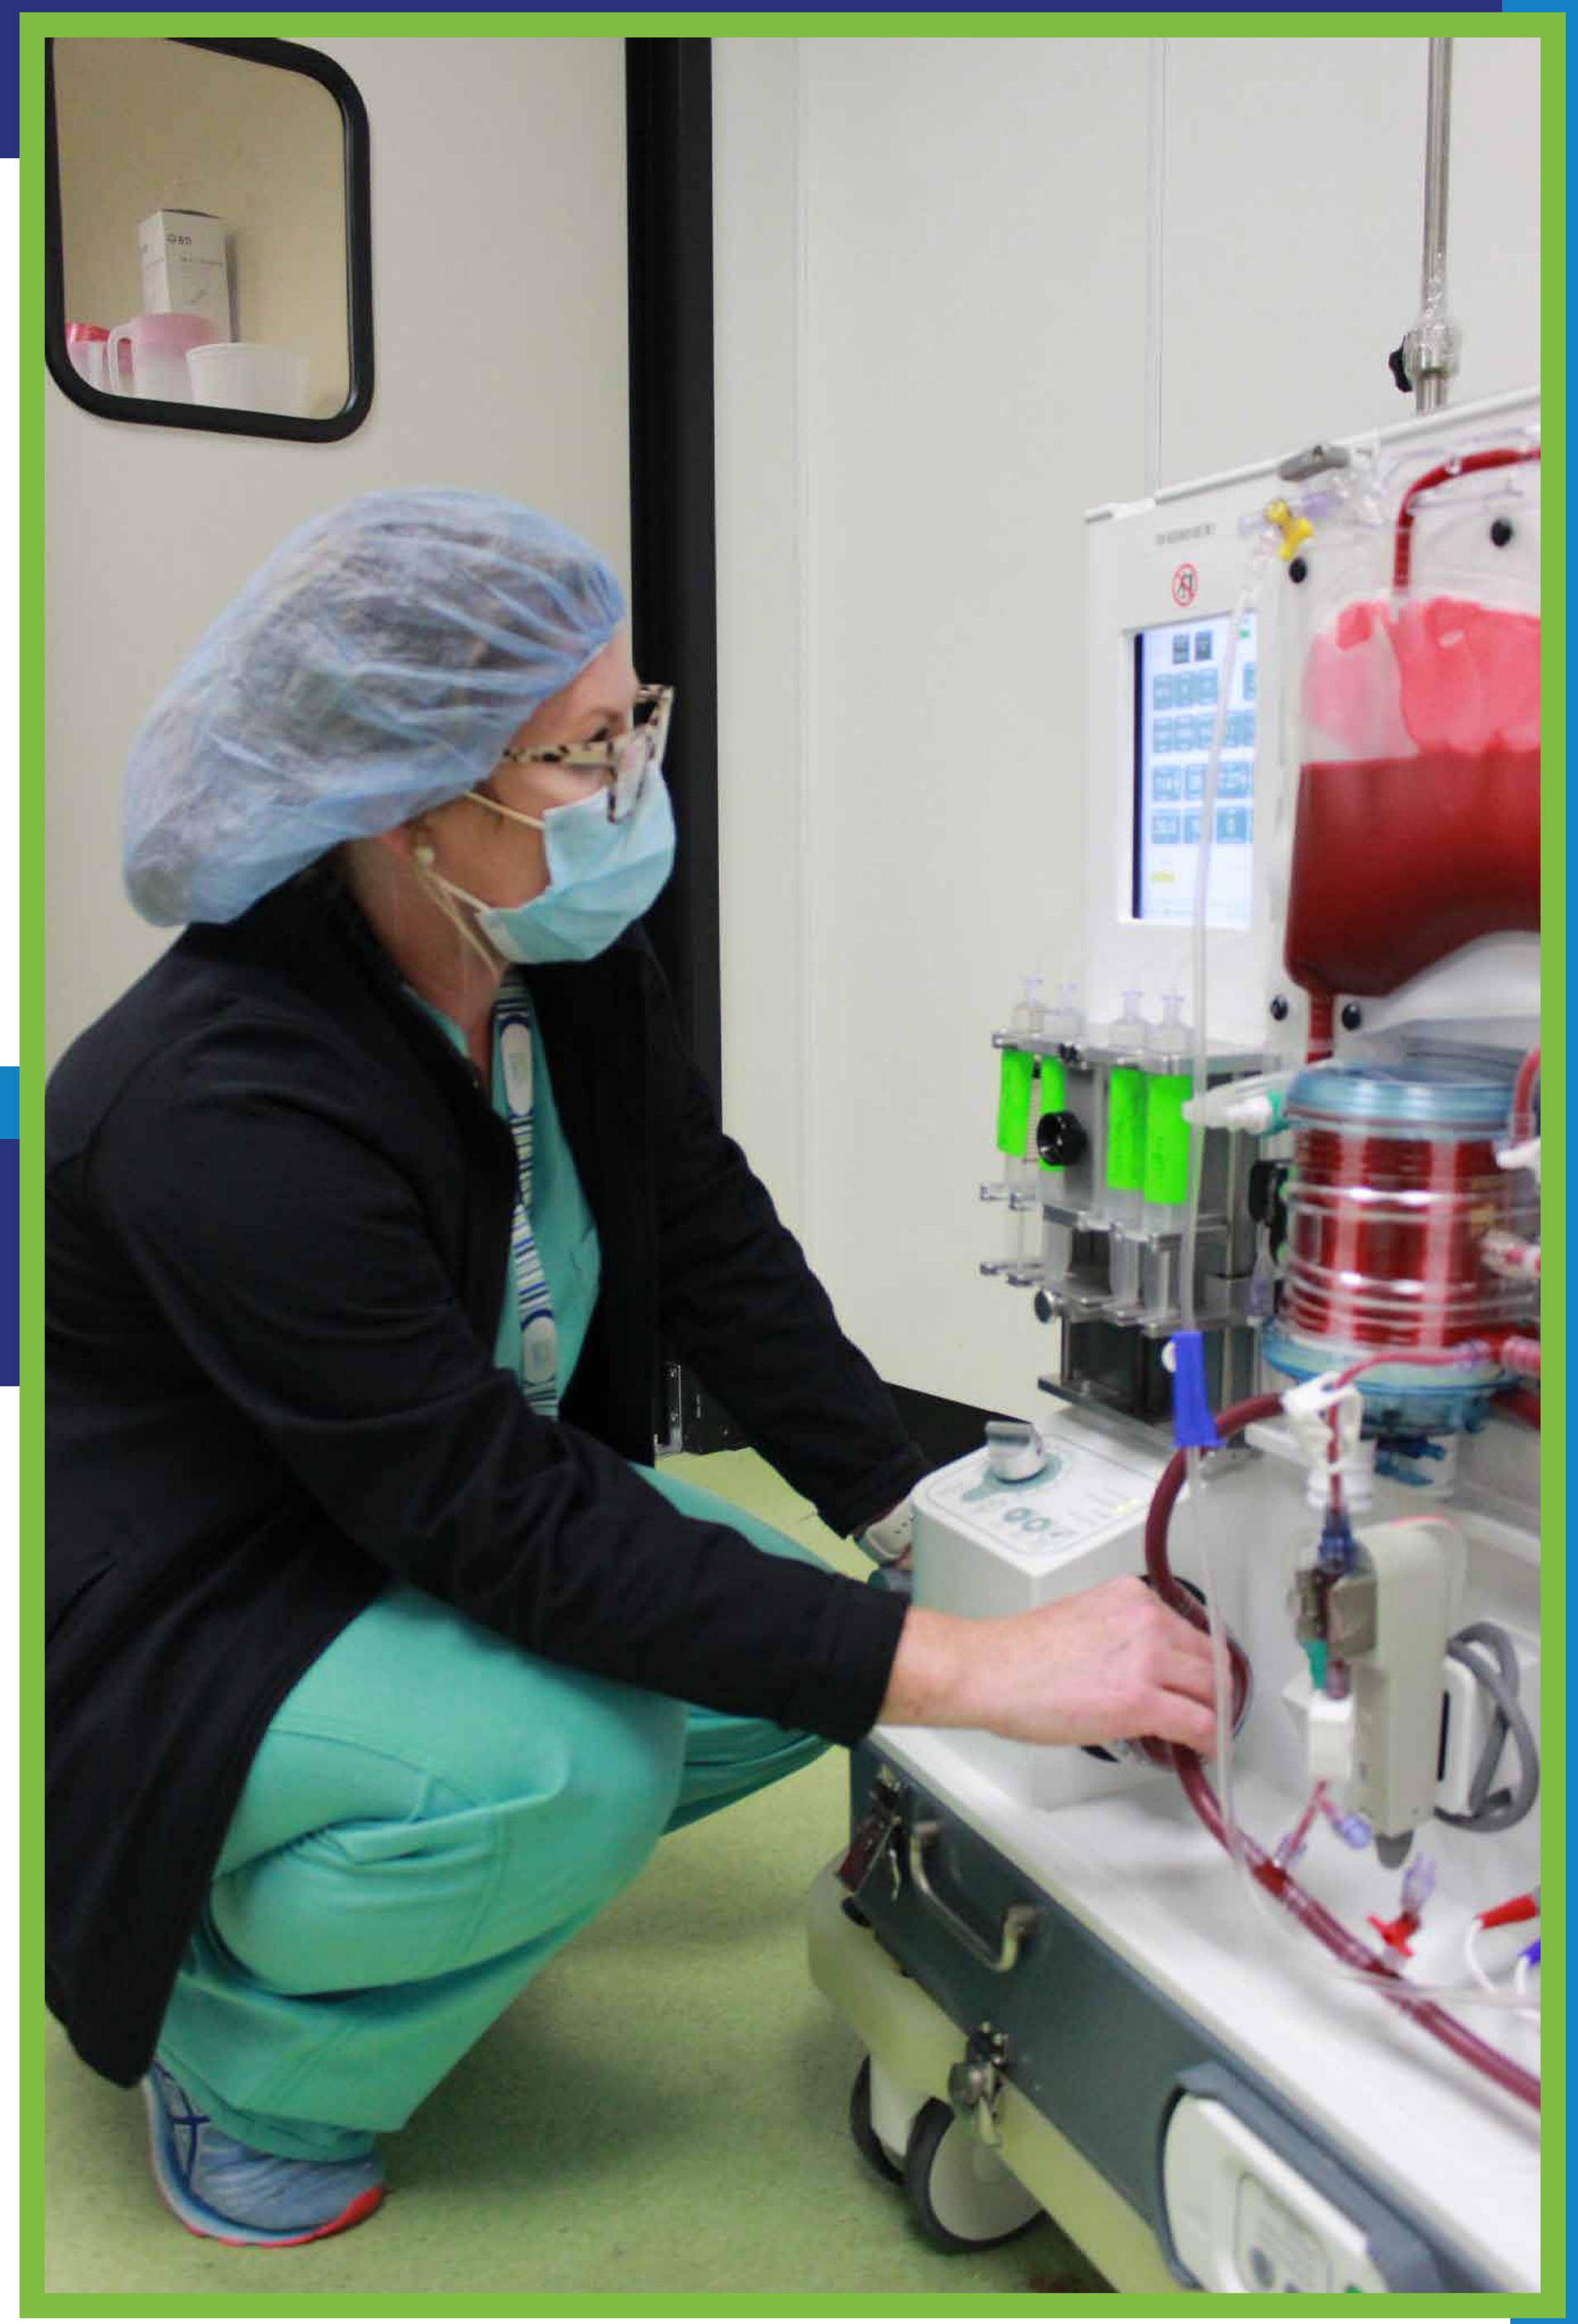

Method

This retrospective case review describes the successful transplantation of donation after brain death donor (DBD) required expedited recovery due to donor instability. The liver was recovered, outside the perfusing OPO's donor service area (DSA), prior to serological testing being able to be completed and then transported to the perfusing OPO's Clinical Innovation Center (CIC) in Oklahoma City via charter flight. Upon arrival, the liver was cannulated and placed on NMP by the perfusing OPO's surgeon and perfusion team using the OrganOx metra®. During NMP, viability was assessed through continuous monitoring of various metrics and liver functions. The center that had accepted with pump and serology waivers participated in real-time ex vivo functional assessment of the liver to determine viability for transplantation.

Timeline

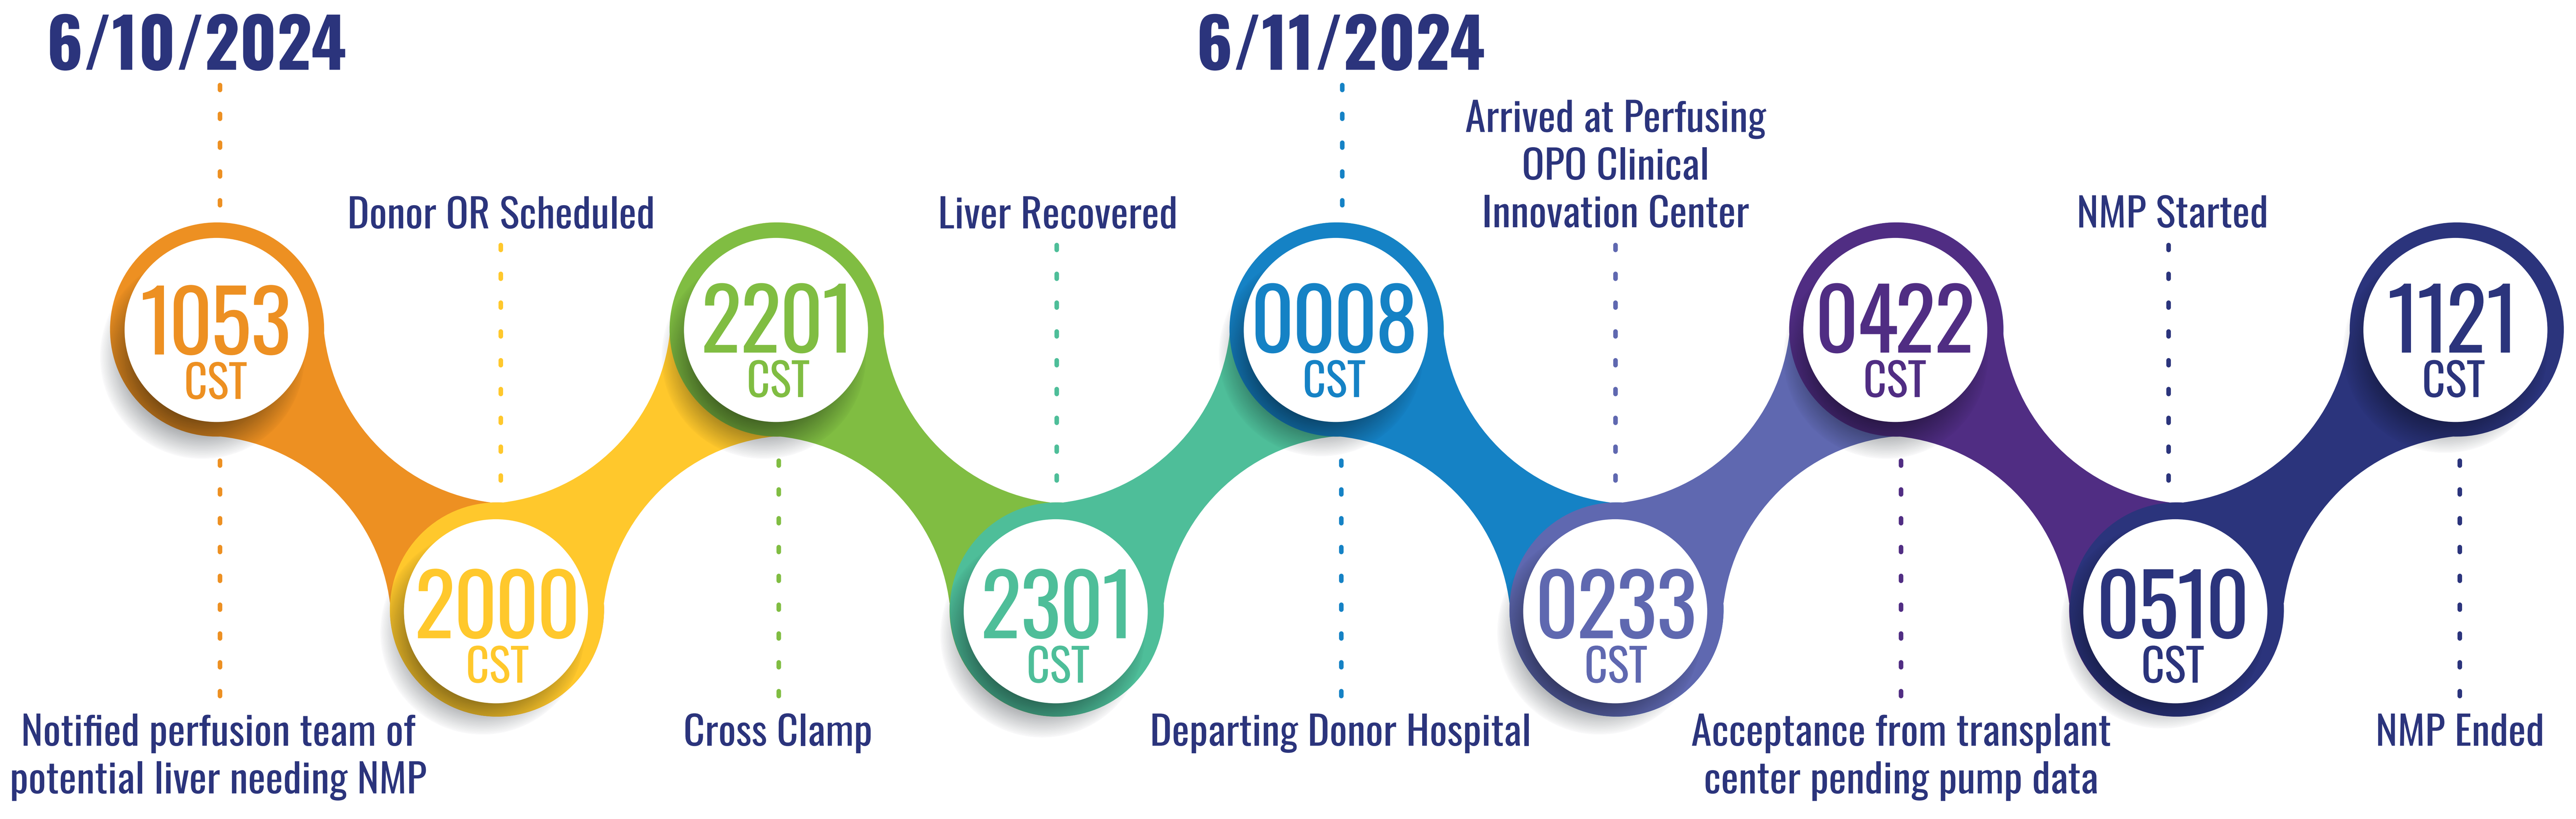

6/10/2024
1053 CST
Notified perfusion team of potential liver needing NMP
2000 CST
Donor OR Scheduled
2201 CST
Cross Clamp
2301 CST
Liver Recovered
6/11/2024
0008 CST
Departing Donor Hospital
0233 CST
Arrived at Perfusing OPO Clinical Innovation Center
0422 CST
Acceptance from transplant center pending pump data
0510 CST
NMP Started
1121 CST
NMP Ended

Results

The cold ischemic time (CIT), from cross clamp at recovery to reperfusion on NMP, was 7 hours 9 minutes. The liver perfusion and assessment took place over a normothermic pump time of 6 hours 11 minutes. The liver cleared lactate to 0.82 mmol/L, maintained a stable pH, metabolized glucose, and produced adequate bile. The functional assessment of the liver resulted in a final acceptance by the transplant center. This liver was transported on NMP by ground to the center where it was ultimately transplanted. The patient was reported to have a normal intraoperative and postoperative hospital course. As of 7 weeks post-op, the liver recipient's labs have been reported within normal limits with no related complications.

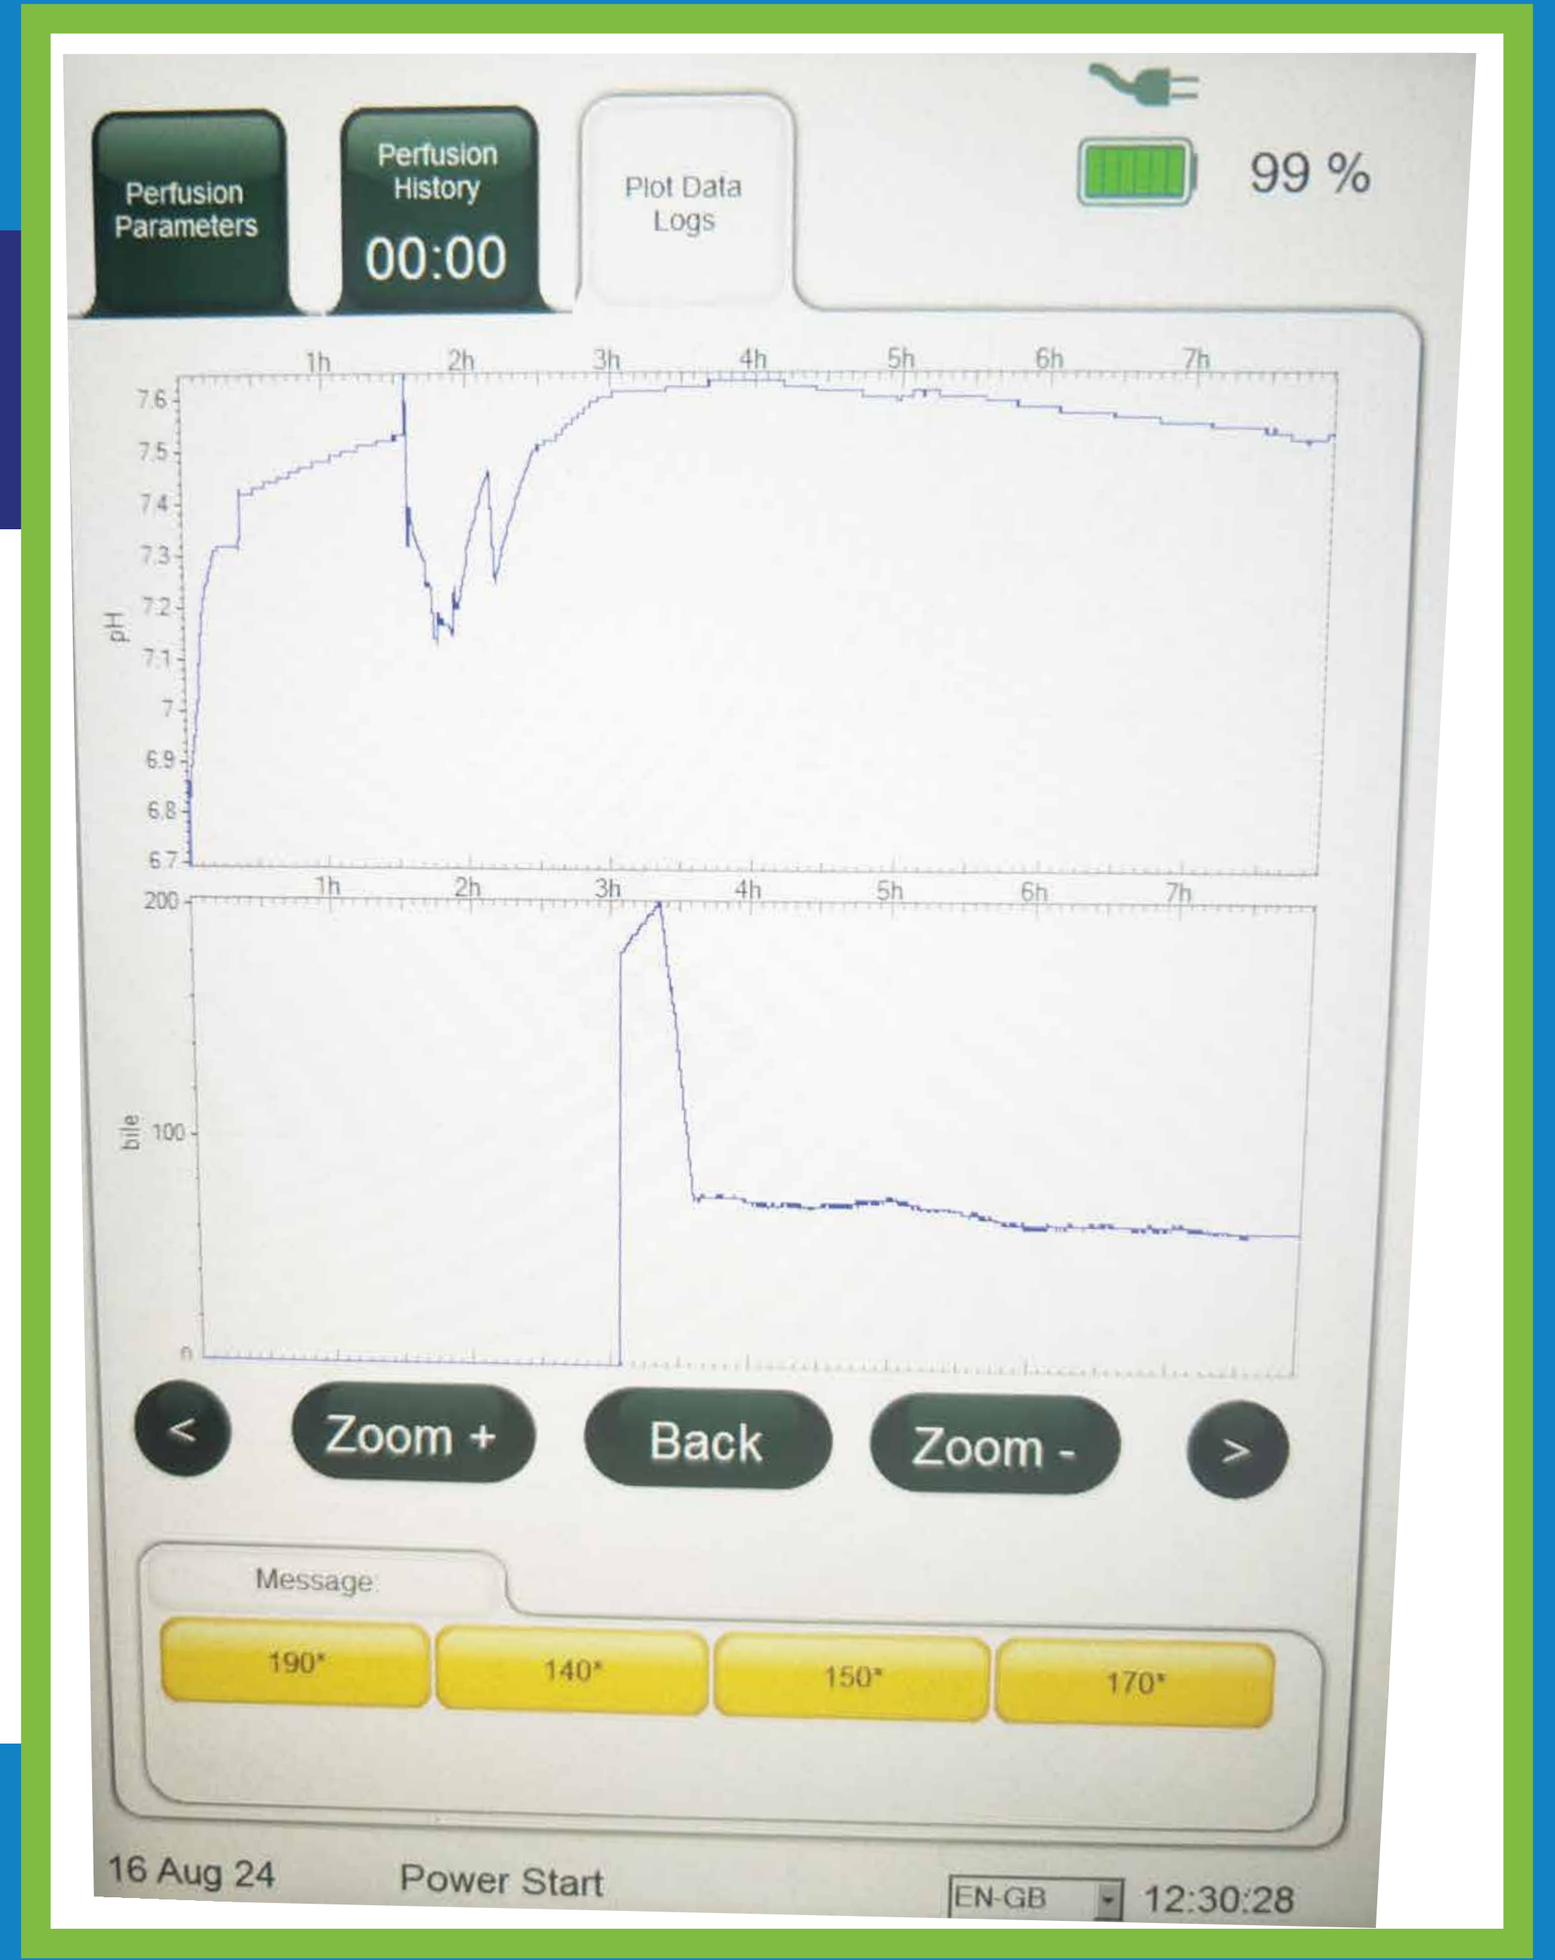

Perfusion Parameters
Perfusion History
00:00
Plot Data Logs
99 %
pH
bile
Zoom +
Back
Zoom -
Message
16 Aug 24
Power Start
EN-GB
12:30:28

Conclusion

We believe this is a model that should be further explored to make NMP more widely available to OPOs who have the opportunity to rescue organs that otherwise are not able to be made available for transplantation. By utilizing NMP and fostering relationships with other OPOs, we can expand the donor pool and rescue livers that might otherwise be discarded due to logistical challenges, resource limitations, or geographical constraints. This innovative approach enhances our ability to maximize organ utilization and improve outcomes for transplant recipients across the nation. OPOs have historically been the place for machine assessment and placement of kidneys, and we believe there are opportunities to replicate this successful model in other extra renal organs.

The authors of the presentation have no conflict of interest or financial relationships related to this abstract.